Efficient Tobramycin Inhalation:

The combination of a high concentration drug with an intelligent inhalation system reduces the initial dosage, the inhalation time and the therapy costs.

Juliane Schick[#], Wolfgang Deml[#], Christine Hardung*, Jens Paulukat[#], Sebastian Canisius[#], Andreas Linke*

[#] Vectura GmbH, Wohraer Straße 37, 35285 Gemünden, Germany; * InfectoPharm GmbH, Von-Humboldt-Str.1, 64646 Heppenheim, Germany

INTRODUCTION

The inhalation of Tobramycin by means of a jet nebuliser generated aerosol is a recognised standard therapy in cystic fibrosis. The pharmacological aim is the highest possible drug deposition in the lower airways and the alveoli. In addition, an acceptable inhalation time is relevant for therapy adherence. Both criteria are influenced by the efficiency of the inhalation system and the concentration (volume) of the Tobramycin solution.

Many nebulisers are inefficient by function or design: an artificial (eFlow® rapid) or an required (jet nebulisers) residual volume remains unused at the end of nebulisation. In addition, there is no automatic way of stopping the nebulisation during expiration which also reduces the amount of the active substance capable of being deposited. Thus, only 10 – 15 % of the initial dosage is deposited in the lungs. A large proportion is wasted, that is, remains unnebulised in the device (residual volume) or is nebulised into the environment; an unnecessary waste and cost to the healthcare system.

According to earlier in-vivo studies from 2006, the AKITA JET® Inhalation System is functionally more efficient. With the development stage of the device and a Tobramycin dose reduced to 160 mg (GERNEBCIN® 160 mg/4 ml), a lung dosage equivalent to 300 mg (TOBI® 300 mg/5 ml) was shown by gamma scintigraphy to be deposited after 13.9 minutes of nebulisation.[1]

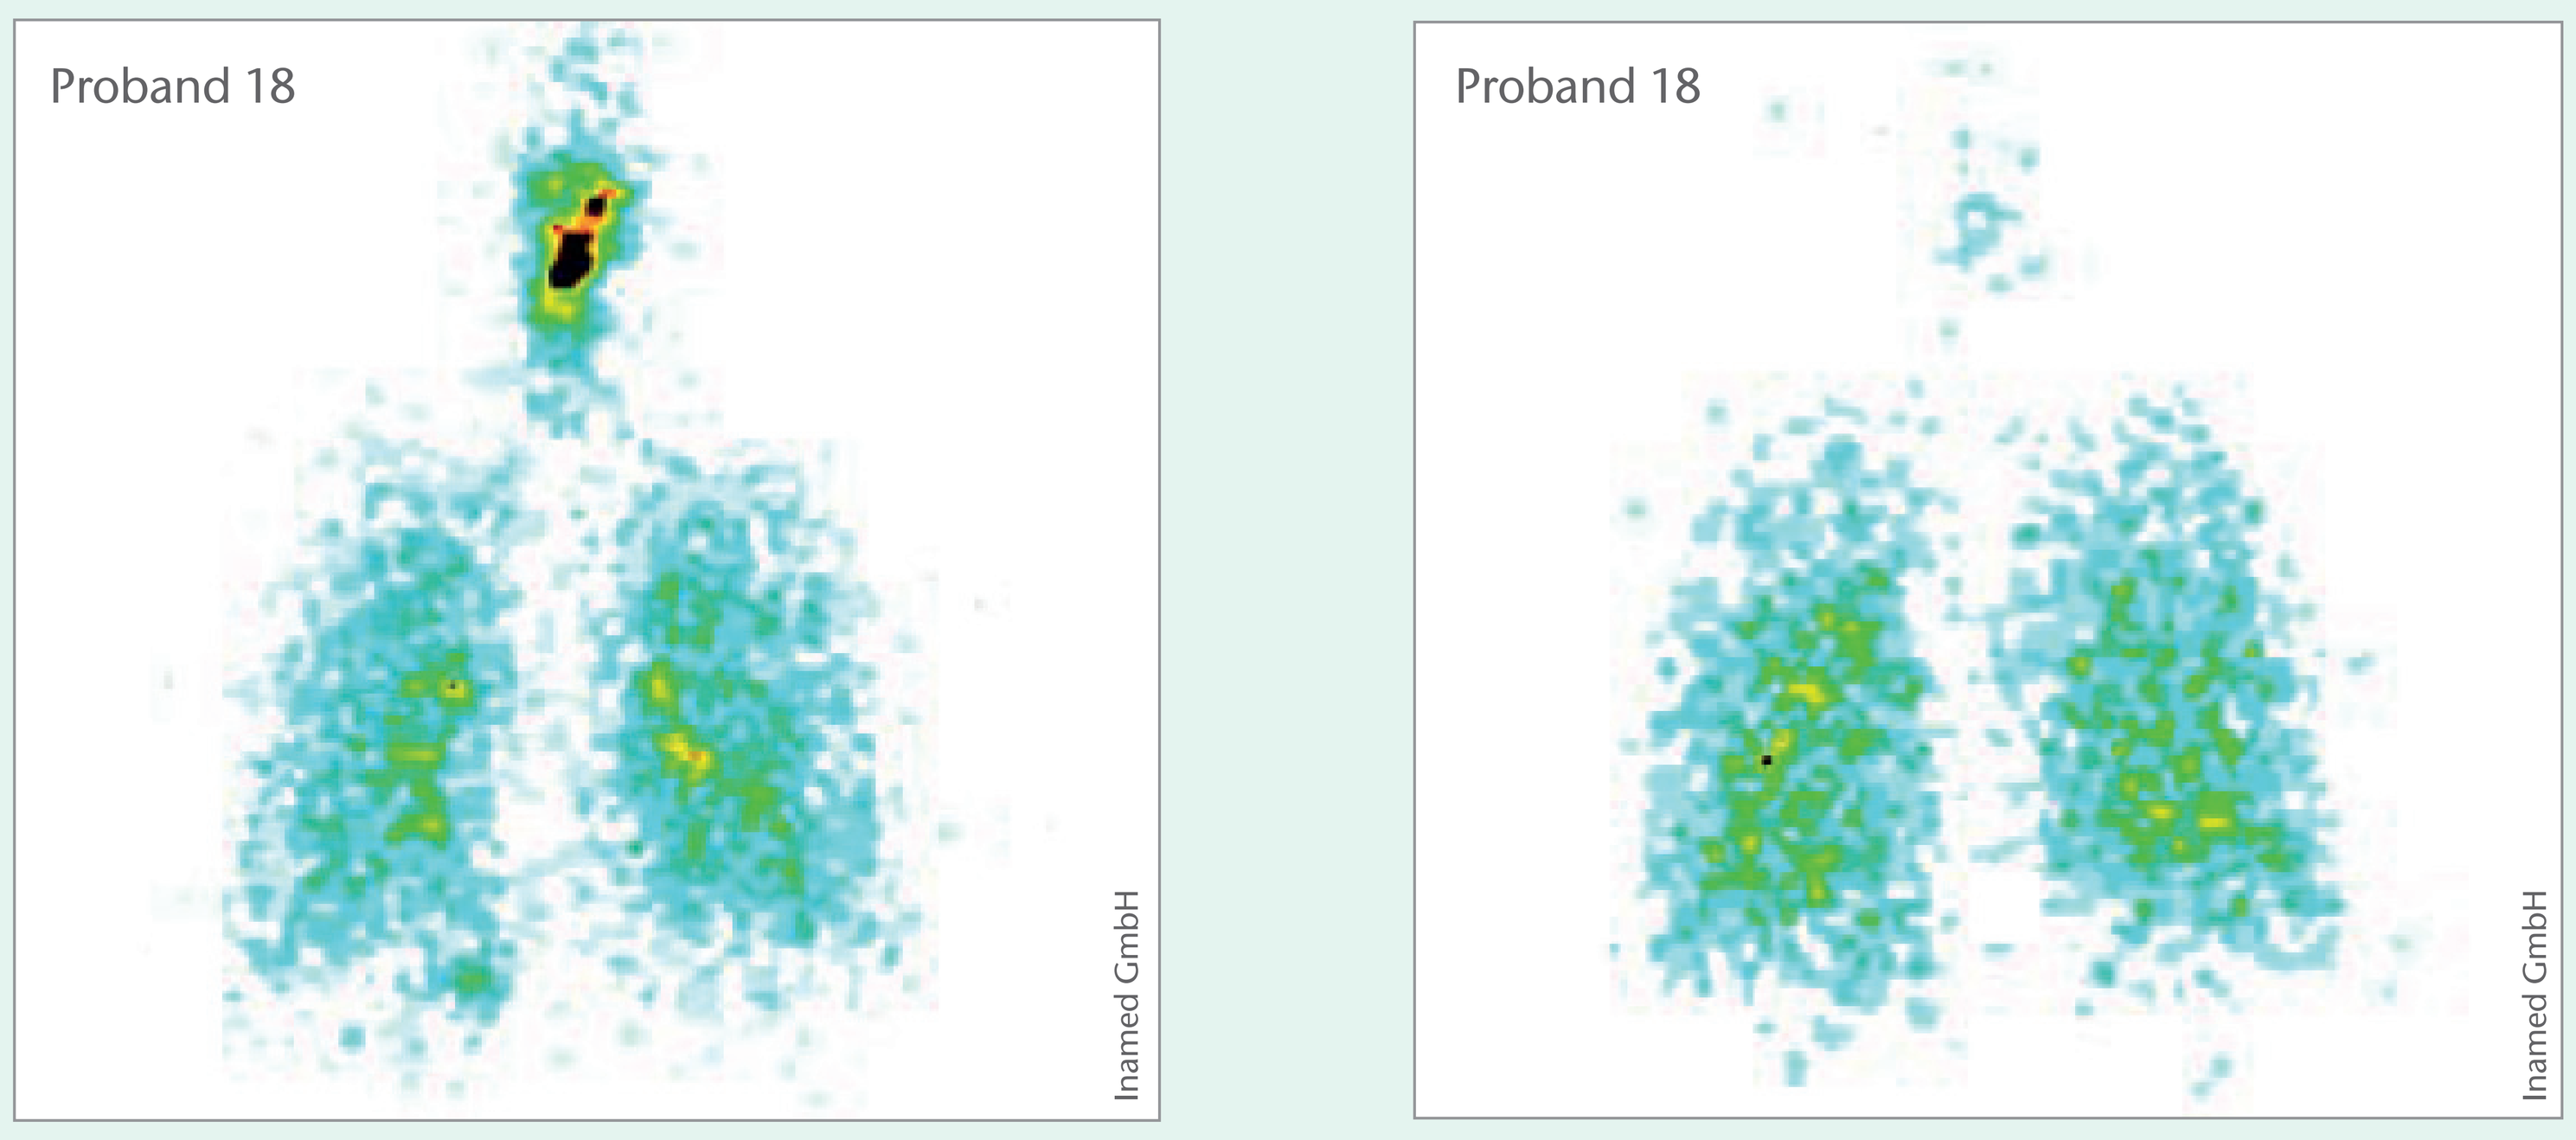
Proband 18
Inamed GmbH
Proband 18
Inamed GmbH

TOBI® 300 mg/5 ml +
PARI boy N, LC Plus®
Lung deposition: 41.8 mg Tobramycin

GERNEBCIN® 160 mg/4 ml +
AKITA JET®, LC Star®
Lung deposition: 43.9 mg Tobramycin

Abb 1: Tobramycin lung deposition recorded by gamma scintigraphy (2006)[1]

STUDY AIM

The aim of the study was to examine whether the new, high concentration GERNEBCIN® 160 mg/2 ml and the current AKITA JET®-system would be able to achieve a lung deposition equivalent to 300 mg TOBI®. The duration of the inhalation should be reduced to under 10 minutes.

METHODS

Two combinations were compared, namely: GERNEBCIN® 160 mg/2 ml + AKITA JET® and TOBI® (300 mg/5 ml) + PARI eFlow® rapid. The aerosols were analysed by laser diffraction (SympaTec Helos; n = 5), and then the active substance proportions in the aerosol as well as in the residual volume were quantified via HPLC-RID (n = 3). A sinusoidal standard adult breathing pattern after DIN EN13544 was generated for the eFlow® rapid (0.5 l/breath; 15 breaths/min).[2] The AKITA JET®-system generates a flow of (200 ml/s) by itself. A typical inhalation volume of 1000 ml was selected (800 ml aerosol + air/200 ml air). The calculation of the lung deposition followed the established ICRP-Lung Model.[3,4]

RESULTS

The fine particle fraction (FPF; ≤ 5 µm) for both systems was over 70 %. In comparison, the AKITA JET® generated smaller particles but a wider range. The delivered Tobramycin dose (DD) was 110.3 mg utilising the TOBI® + eFlow® rapid combination. Despite the almost double initial dose, this was only 1.5 times higher than that generated by the AKITA JET®-system with GERNEBCIN® (73.0 mg). In contrast to the AKITA JET®, the eFlow® rapid also delivered 75.0 mg Tobramycin into the environment (AL) during the expiration cycle due to its continuous nebulisation function.

Following the ICRP-model, it was calculated that both systems delivered a comparable total lung dose (LD) of 50.8 mg (eFlow® rapid) and 53.6 mg (AKITA JET®), respectively. There were differences in the pattern of deposition. The AKITA JET® deposited 64% (34.2 mg) oft the lung dosage in the remote peripheral-alveolial areas of the lung, whereas the eFlow® rapid managed this with only 51 % of the LD (26.1 mg).

The eFlow® rapid nebulised TOBI® in only 5.8 min. The inhalation time needed by the actual AKITA JET®-GERNEBCIN® combination amounted to 9.7 min. This is markedly shorter than that recorded in 2006 (13.9 min).

SUMMARY

The AKITA JET® Inhalation System generated an equivalent lung dose to 300 mg TOBI® with only 160 mg GERNEBCIN® in less than 10 minutes. In addition, a significant amount of the aerosol was delivered to the remote peripheral-alveolial areas. Due to the smaller initial dosage and the utilisation of the AKITA JET®, the daily therapy costs could be reduced by 68.64 €/day (43 %) .

The superior efficiency of the AKITA JET® is due to the technology (FAVORITE*), that actively leads and supports the patient's breathing: A constant flow of 200 ml/s ensures a high pulmonary deposition rate and an air bolus at the end of each inspiration ensures a deposition deep in the lungs. A cessation of the nebulisation during the expiration phase eliminated aerosol loss to the atmosphere.

The eFlow® rapid is the quicker system. It nebulises 5 ml TOBI® in just 6 minutes. Nevertheless, in comparison to the earlier study the inhalation time for GERNEBCIN® with the AKITA JET® was reduced by 30%. The actual duration of only 9.7 minutes reflects the higher active substance concentration in GERNEBCIN® 160 mg/2 ml.

	VMD [µm]		GSD		FPF [%]	
	Mean	SD	Mean	SD	Mean	SD
GERNEBCIN® 160 mg/2 ml	3.57	0.01	1.95	0.01	71.46	0.14
TOBI® 300 mg/5 ml	3.93	0.04	1.5	0.01	73.75	1.13

VMD: volume mediane diameter; GSD: geometric standard deviation; FPF: fine particle fraction ≦ 5 µm

Tab 1: Aerosol-characterisation by laser diffraction

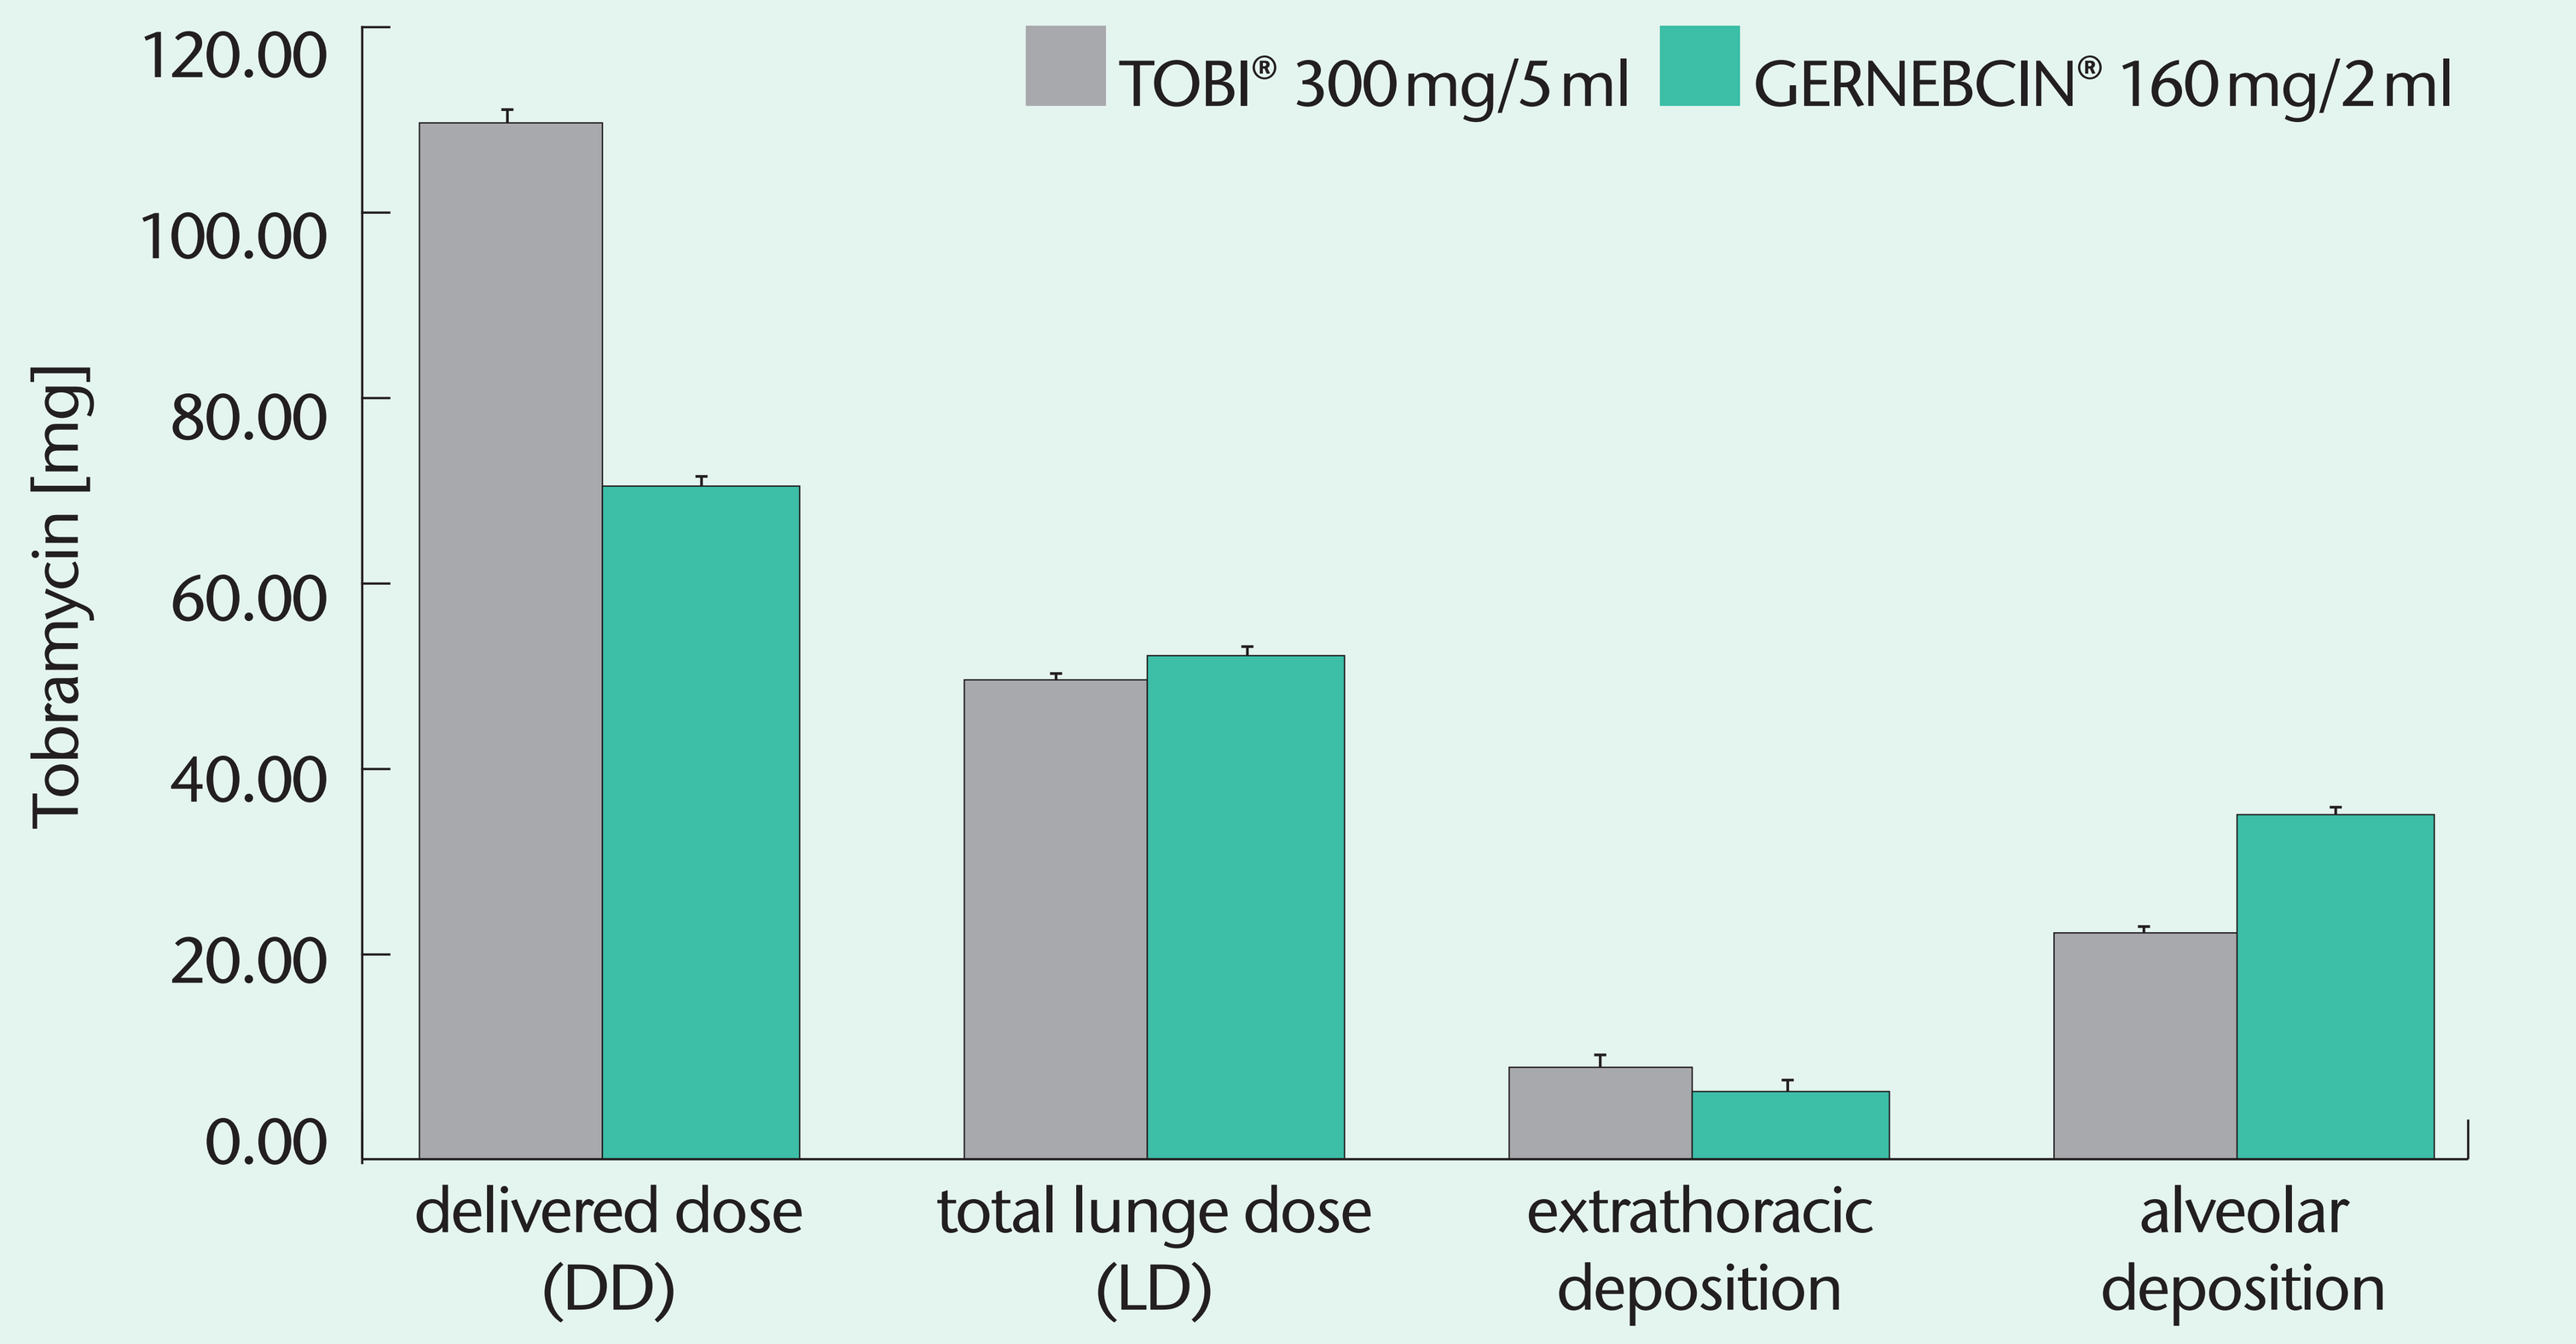
120.00
100.00
80.00
60.00
40.00
20.00
0.00
Tobramycin [mg]
TOBI® 300 mg/5 ml
GERNEBCIN® 160 mg/2 ml
delivered dose (DD)
total lunge dose (LD)
extrathoracic deposition
alveolar deposition

Abb 2: Comparison of the delivered dose (DD) with the dose deposited in the lung (LD) according to ICRP[3,4], the alveolar deposited proportion and the extrathoracic loss

Drug	TOBI® 300 mg/5 ml		GERNEBCIN® 160 mg/2 ml	
Inhalation system	PARI eFlow rapid		AKITA JET/LC Sprint	
	Mean	SD	Mean	SD
Delivered dose of Tobramycin (DD) [mg]	110.33	1.89	73.00	2.00
Aerosol loss to the atmosphere (AL) [mg]	75.00	2.00	0.00	0.00
Residual dose of Tobramycin (DR) [mg]	100.08	3.88	91.17	2.65
Lung dose of Tobramycin (LD) [mg]	50.77	0.87	53.55	1.47
Lung dose in relation to initial dose [%]	16.92	–	33.47	–
Time of inhalation [min]	5.83	0.11	9.67	0.00
Current therapy costs/day, inhalation twice daily [€][5]	119.94	–	51.30	–

Tab 2: Overview of study results including a comparison of daily therapy costs[5]

LITERATURE

1 Brand et al. 2006: Reduction of drug-dose and therapy-costs in the inhalation therapy with high dose tobramycin, Journal of Cystic Fibrosis 2006;Vol 5:S40

2 EN13544-1: 2007+ A1:2009: Atemtherapiegeräte – Teil 1: Verneblersysteme und deren Bauteile

3 Fleming et al. 2006: Comparison of SPECT aerosol deposition data with a human respiratory tract model, Journal of Aerosol Medicine 2006;Vol 19(3):268-78

4 ICRP 1994: Human respiratory tract model for radiological protection. A report of a Task Group of the International Commission on Radiological Protection. Ann ICRP 1994;Vol 24(1-3):1-482

5 pharmacy retail price (ABDA-database; www.pharmazie.com; 28.08.2014)

* FAVORITE: Flow and Volume Regulated Inhalation Technology

contact: andreas.linke@infectopharm.com

S8000251-01-0115